Cannabidiol based borderline products - from development to registration processes

Aleksandrov M.*[1,2], Gelevska O.[1], Stojova C.[1], Ivanov I.[1], Ananieva Bozinov L.[1], Maksimova V.[2]

[1]TetraHip LLC Kocani, Orizova 9, 2300 Kocani, Republic of North Macedonia

[2]Faculty of Medical Sciences, Goce Delcev University of Stip, Krste Misirkov 10A, 2000 Stip, Republic of North Macedonia

INTRODUCTION

- ✓ Cannabidiol (CBD) is the second most abundant phytocannabinoid structure in *Cannabis sativa* L., Cannabaceae family.
- ✓ Cannabidiol based products have been used mainly for additional treatment of pain, depression and anxiety in cancer suffering patients as well as immunostimulatory agents.
- ✓ Pharmacological effects are exerted through the endocannabinoid system through multiple receptors, including cannabinoid receptors (CBRs), opioid and serotonin receptors, adrenergic receptors, and G protein-coupled receptors.
- ✓ A borderline product may be a product that contains active components of plant origin, and it is classified as a food supplement in at least one member state of the European Union, and according to the special regulations applied in the Republic of North Macedonia, it cannot be classified as a food supplement; prescribed in accordance with the special regulations, if it is classified as a food supplement in at least one member state of the European Union and there is no registered product with the same strength in the Republic of North Macedonia (Official Gazette of R.M no. 203/2015).
- ✓ **According to the second term of this law CBD products are still registered as borderline products in our country.**

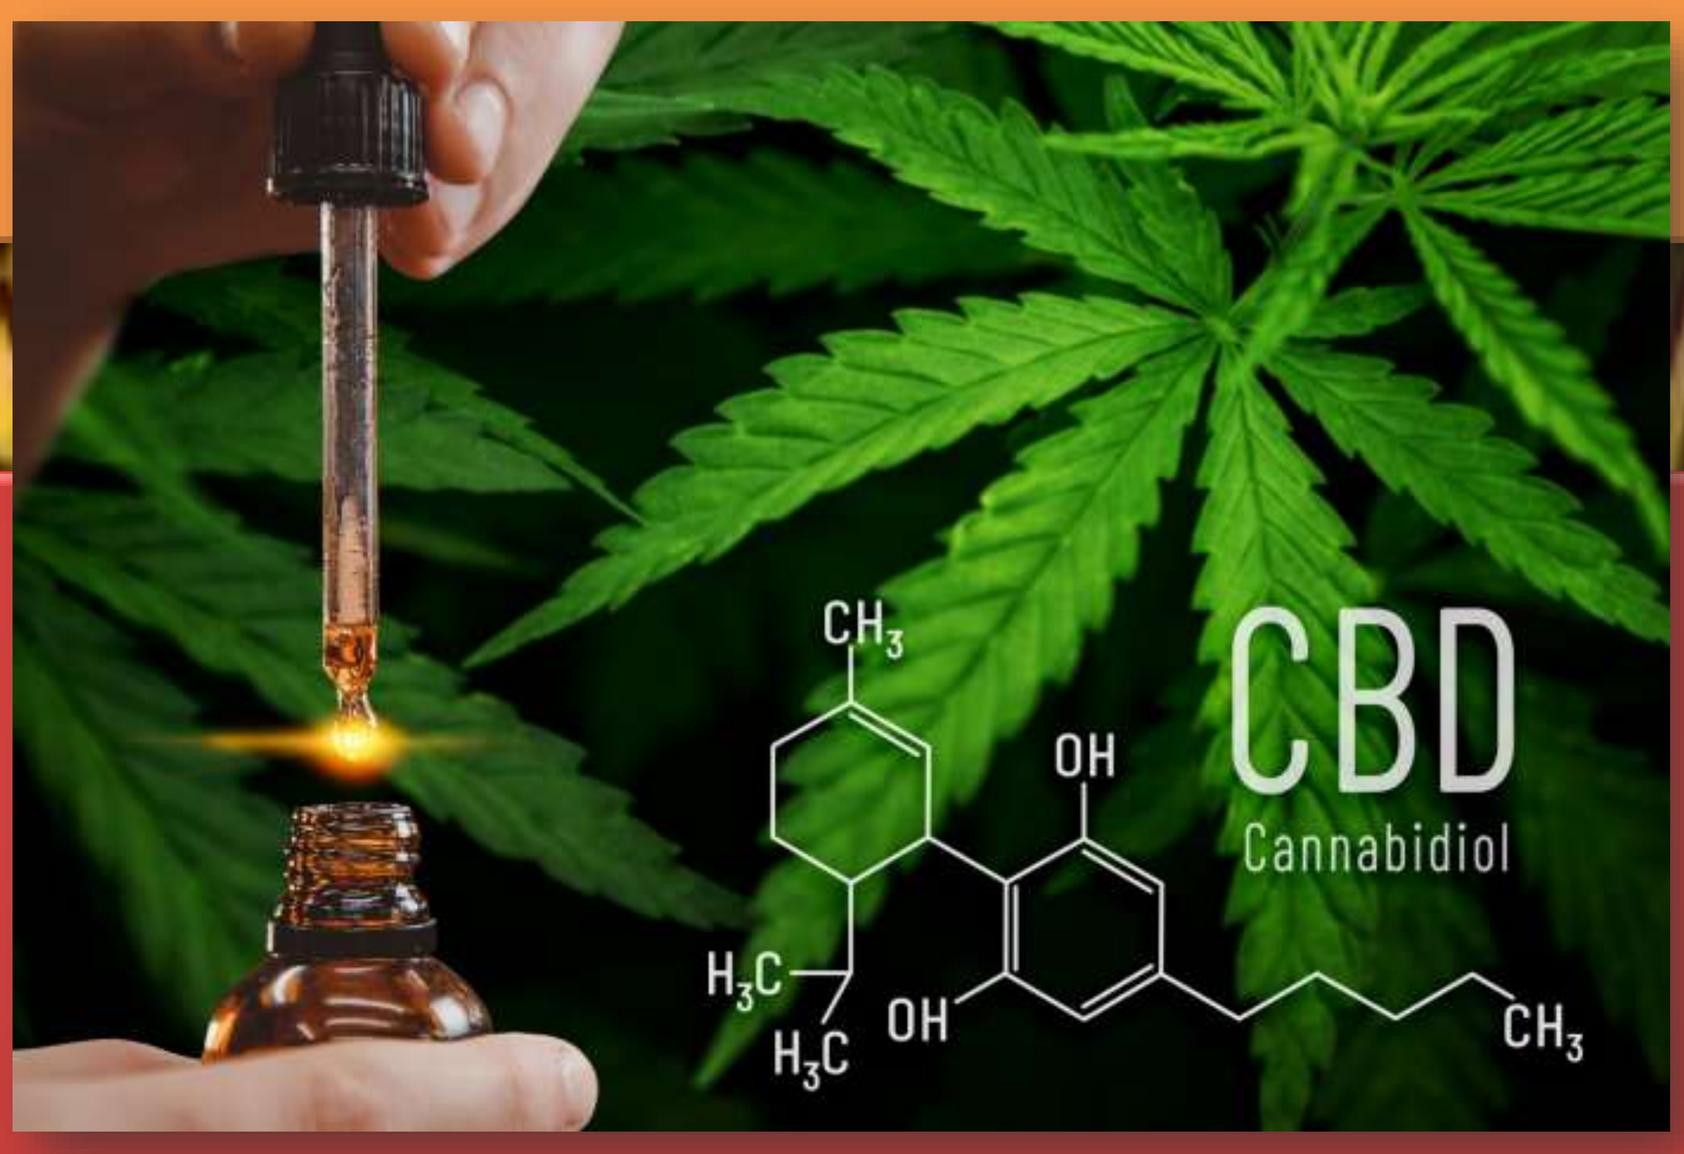
CBD
Cannabidiol
CH_3
OH
H_3C
H_3C
OH
CH_3

MATERIALS AND METHODS

- *dry hemp biomass* (DHBM) (obtained from Bioherbalist, Czech Republic);
- *medium-chain triglycerides oil* (MCT oil) Type V, Ph. Eur.10.0 (obtained from GustavHeess, Germany);
- *hemp seed oil, refined* (HSOr) (obtained from GustavHeess, Germany);
- *ethanol 96% v/v* Ph. Eur. 10.0 (EtOH) (obtained from Alkaloid AD, North Macedonia).
- All materials were provided with certificates of quality from the supplier, according to the pharmacopeial requirements.

The **ethanol extraction (EE) line** consist of chillers, solvent extractors, centrifugal filters, buffer filtering equipment (BFE), falling film concentration equipment (FFCE), vacuum spherical concentration equipment (VSCE), filter separator, vacuum pumps and solvent recovery system.

Ethanol extraction of the DH-BM was performed by appropriate EE line. First, a cold maceration of the DH-MB was performed with cold ethanol, with a ratio of DH-BM: EtOH=1:10 (w/v) for 1 h.

After centrifugation of the mixture, ethanol extract was filtered and placed in **thin-layer (film) distillation** took in the FFCE at temperature of 78°C.

By these methods, the ethanol extract was obtained and afterward filtered through BFE. Concentration and ethanol evaporation of the extract were performed in VSCE, at the temperature of 80°C with homogenization. By these procedures, crude cannabis oil (CCO) was obtained.

Decarboxylation of CCO was performed in Double Jacketed Glass Reactor at a temperature of 140°C, followed by a **short path distillation** in order to purify and separate cannabinoids in cannabis extract.

Production of oil solutions – oil solutions of standardized cannabis extract (SE-CBD) were prepared in two types: (1)oil solutions in MCT oil and (2)oil solutions in HSOr. The mixing of the oil solution took place in a magnetic stirrer at a temperature of 65ºC with 200 rpm for 45 minutes.

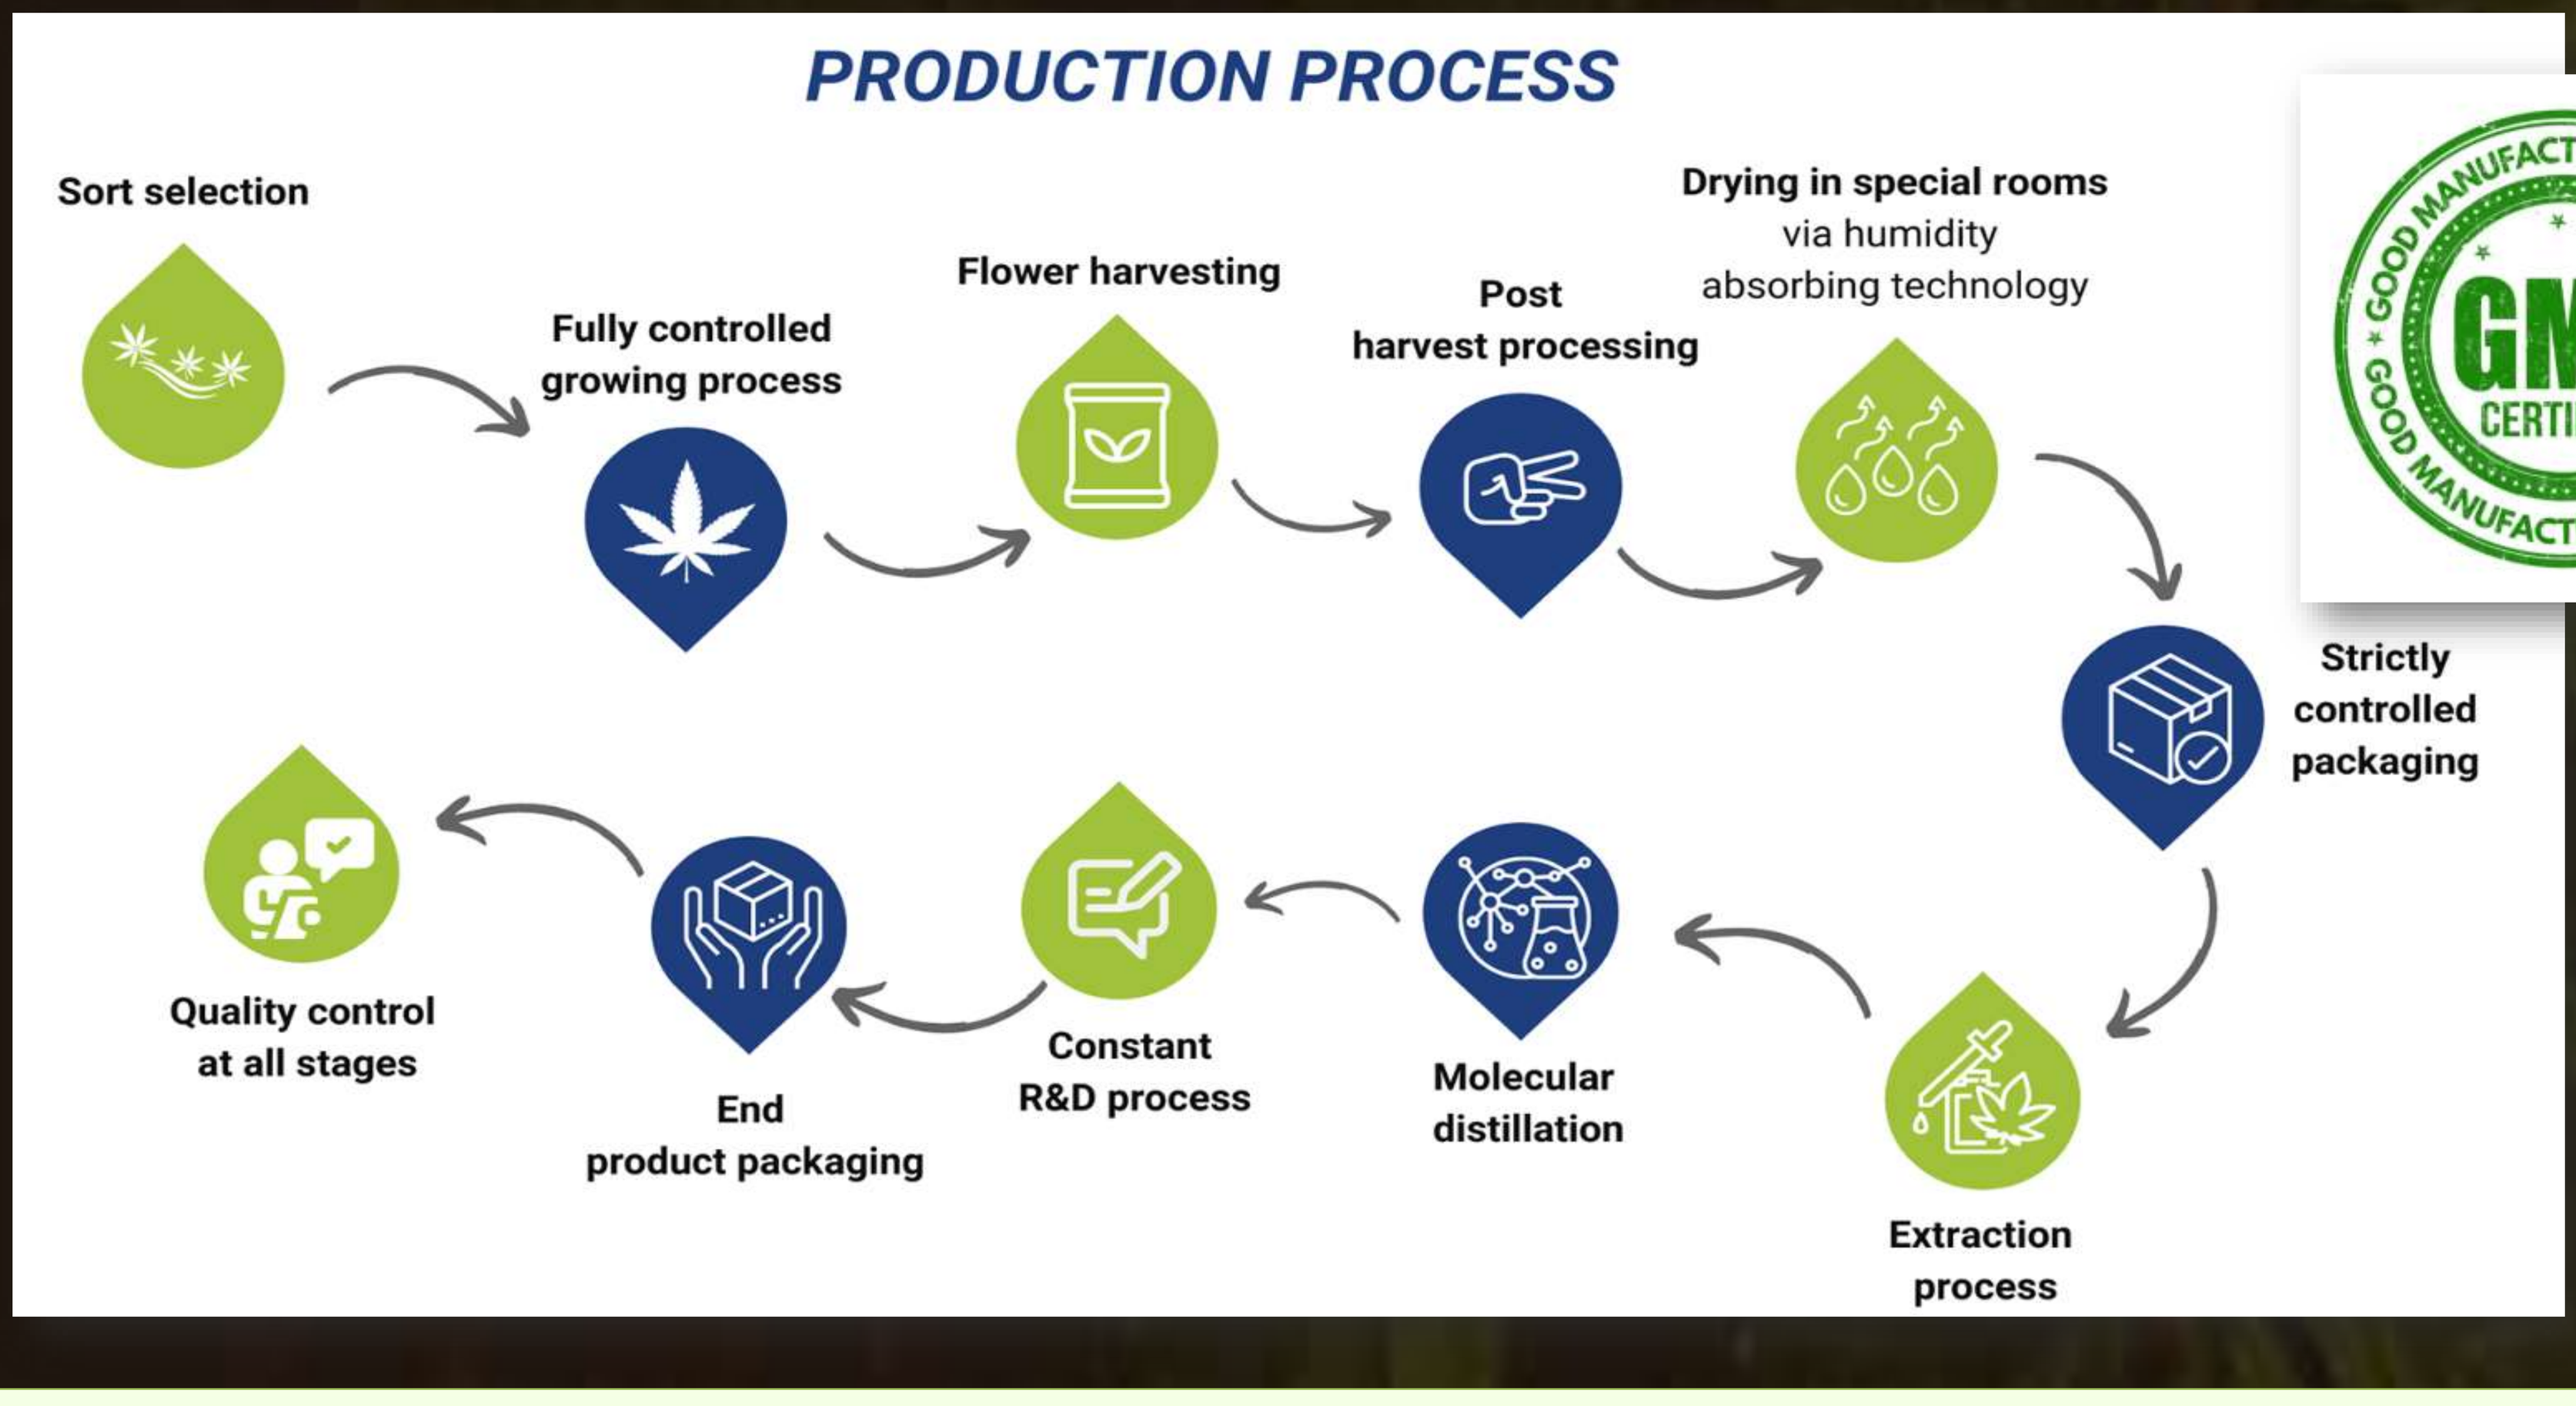
PRODUCTION PROCESS
Sort selection
Fully controlled growing process
Flower harvesting
Post harvest processing
Drying in special rooms via humidity absorbing technology
Strictly controlled packaging
Extraction process
Molecular distillation
Constant R&D process
End product packaging
Quality control at all stages

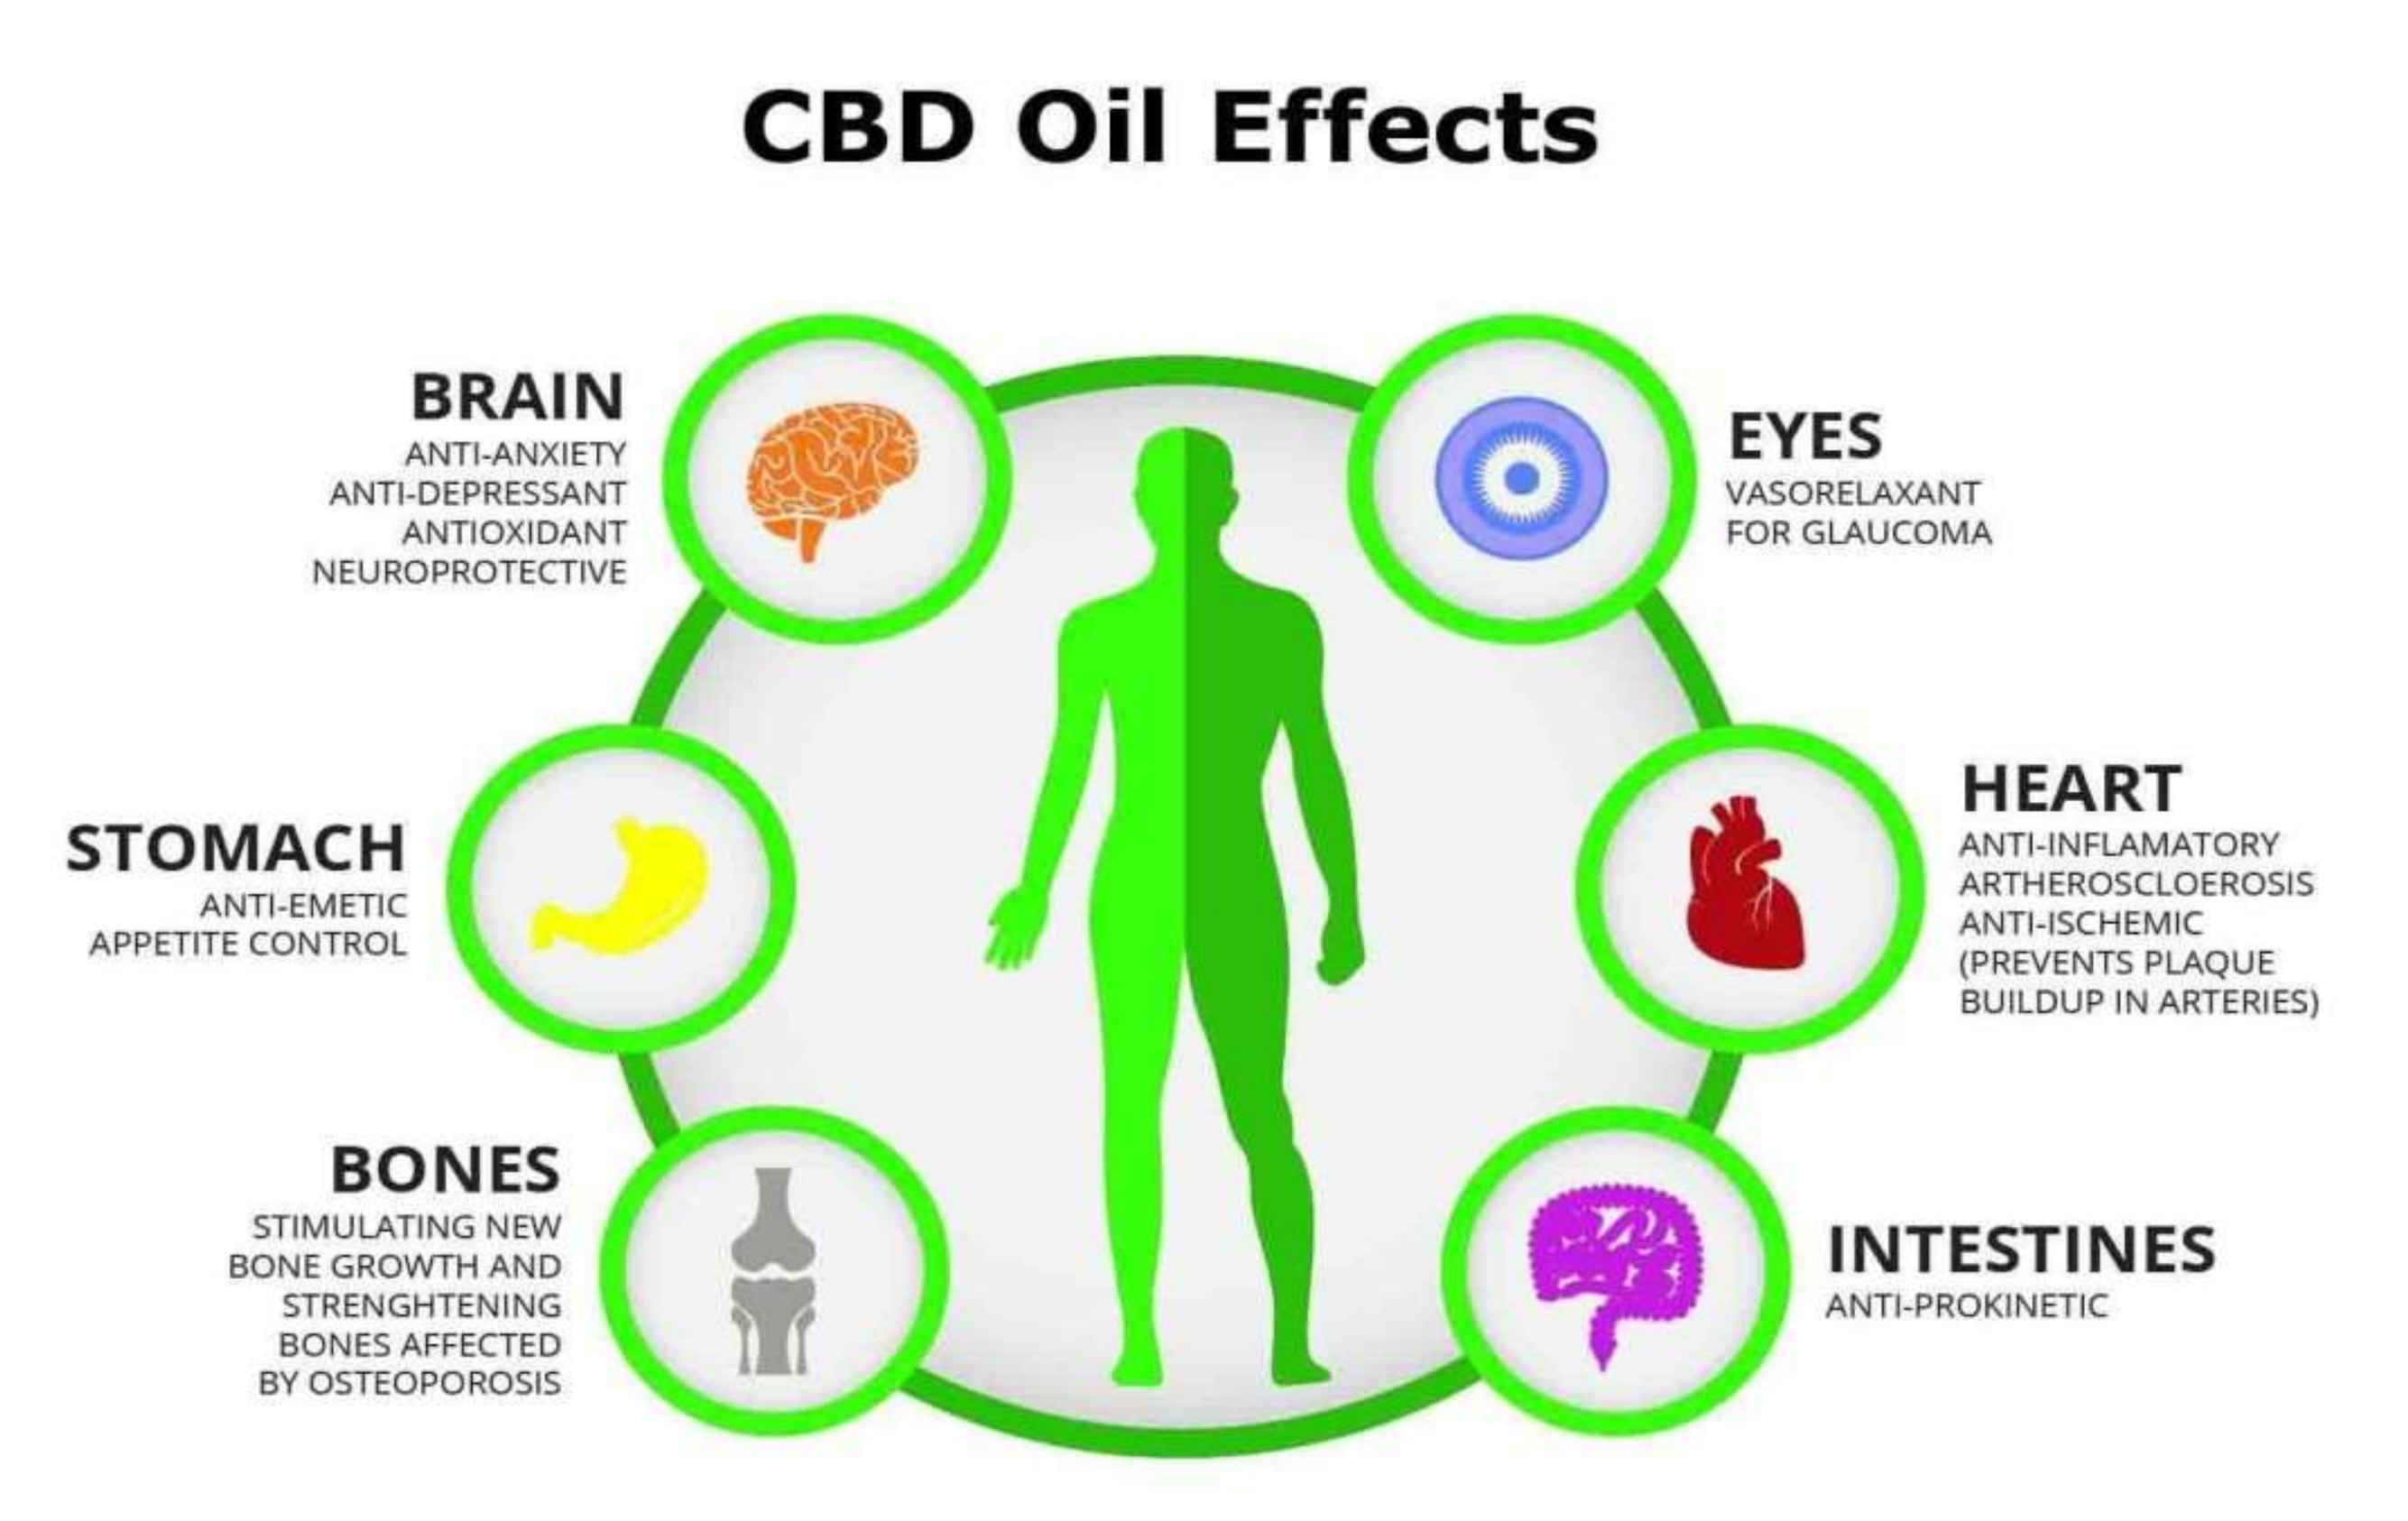
CBD Oil Effects
BRAIN
ANTI-ANXIETY
ANTI-DEPRESSANT
ANTIOXIDANT
NEUROPROTECTIVE
EYES
VASORELAXANT
FOR GLAUCOMA
STOMACH
ANTI-EMETIC
APPETITE CONTROL
HEART
ANTI-INFLAMATORY
ARTHEROSCLOEROSIS
ANTI-ISCHEMIC
(PREVENTS PLAQUE
BUILDUP IN ARTERIES)
BONES
STIMULATING NEW
BONE GROWTH AND
STRENGHTENING
BONES AFFECTED
BY OSTEOPOROSIS
INTESTINES
ANTI-PROKINETIC

RESULTS & DISCUSSION

We have obtained a high-purity and superior quality concentrate that can be processed into a variety of final products.

Standardized extracts SE-CBD contain a total of CBD=70.6% (w/w) and total THC=0.17% (w/w), after in process control (IPC).

All IPC procedures were performed, and the final products were analyzed by a validated HPLC method with UV detector.

Three batches of each of the two types of oil solutions were made.

The first batch contained 5% (*w/w*) CBD, the second batch 10% (*w/w*) CBD and the third batch 20% (*w/w*) CBD. All batches were packed in bulk of 200 ml in dark glass bottles.

The received certificates for physico-chemical and microbiological tests were in accordance with the set specifications for the quality of the finished product.

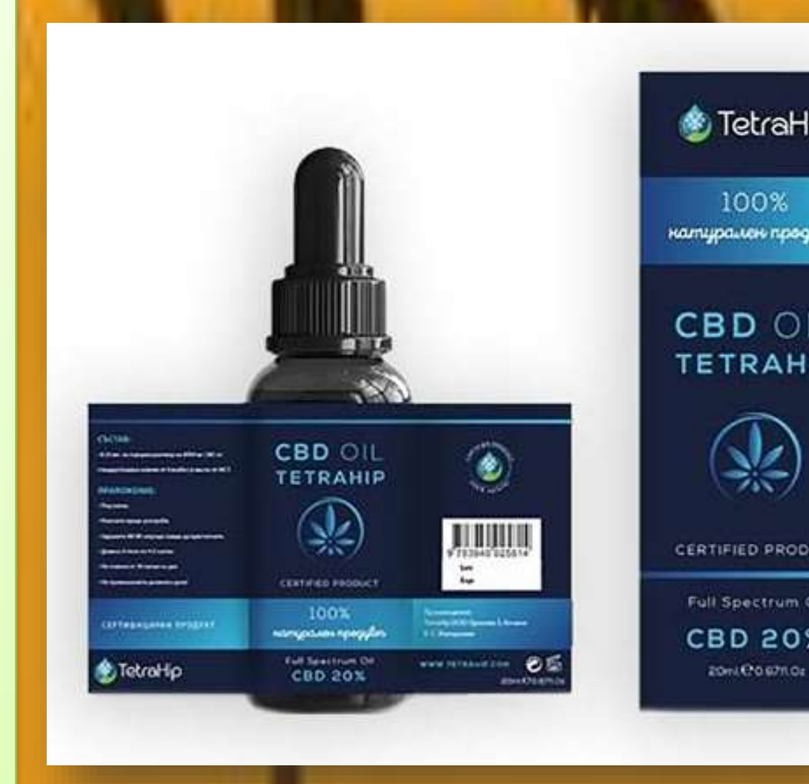
TetraHip
100%
CBD OIL
TETRAHIP
CERTIFIED PRODUCT
Full Spectrum Oil
CBD 20%

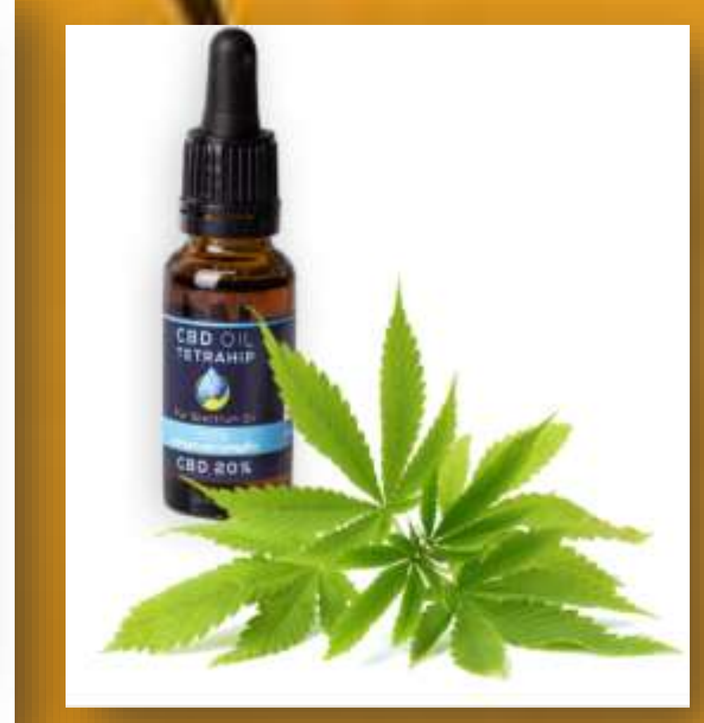

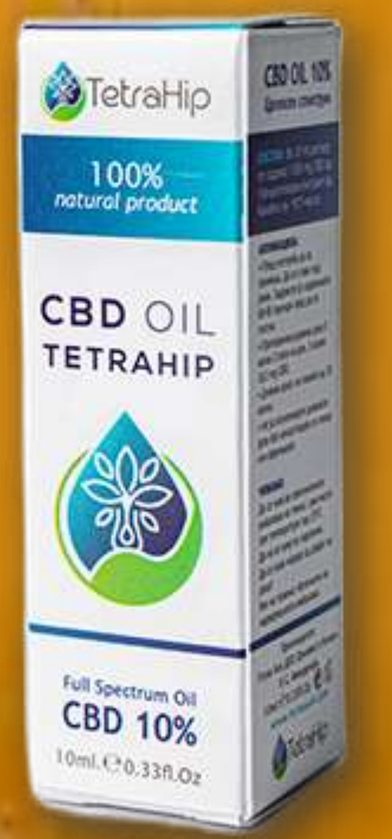
TetraHip
100%
natural product
CBD OIL
TETRAHIP
Full Spectrum Oil
CBD 10%

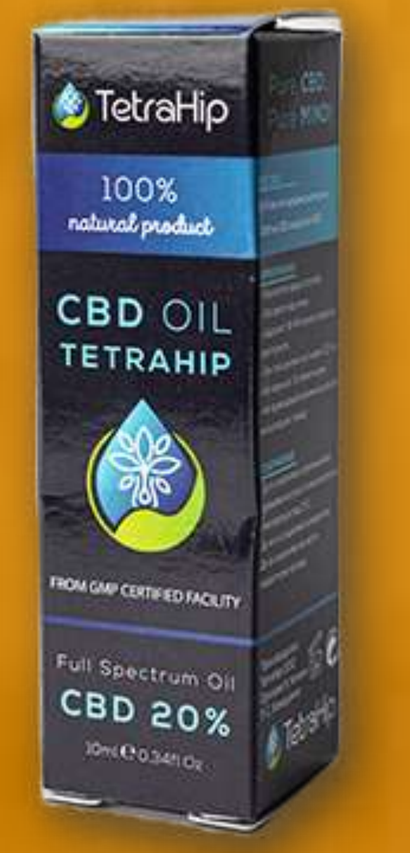
TetraHip
100%
natural product
CBD OIL
TETRAHIP
FROM GMP CERTIFIED FACILITY
Full Spectrum Oil
CBD 20%

Registration procedure for classification of the products into borderline products was initiated

This process was supported with following documentation:

(1)Request for product classification, in which the name of the product, the manufacturer, the country of origin, the agent, or the importer of the product have been specified;

(2)A sample of the product for which classification is requested in final packaging;

(3)Exact qualitative and quantitative composition of the product and certificates for product analysis;

(4)Patient Information Leaflet (PIL) if not an integral part of the product's outer packaging;

(5)List of countries in which the sale of the product is approved with its classification;

(6)Product classification certificate issued by a competent authority from a member state of the European Union;

(7)Free Sales Certificate, and

(8)Negative opinion from AHV if the product comes from abroad and is registered as a dietary supplement.

MALMED

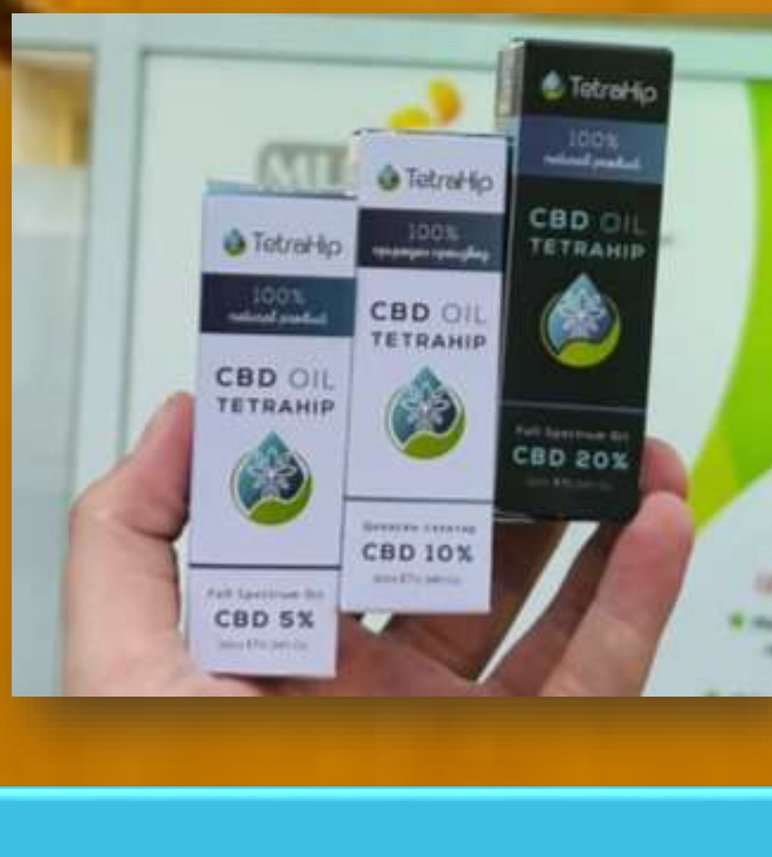
TetraHip
CBD OIL
TETRAHIP
CBD 5%
CBD 10%
CBD 20%

Twelve CBD-based borderline products were registered by MALMED. Six of them contained MCT oil, and the other six were with HSOr, as excipients. Borderline products contain different concentrations of CBD (5% w/w, 10% w/w and 20% w/w) and are packaged in dark glass bottles (with dropper) of 10 ml and 20 ml.

These products can be dispensed and/or sold without a prescription (OTC).

Sublingual administration allows direct absorption of CBD and faster delivery of higher concentrations of CBD into the systemic circulation. Another advantage of this route administration is the possibility of easier dosing by gradually increasing the dose.

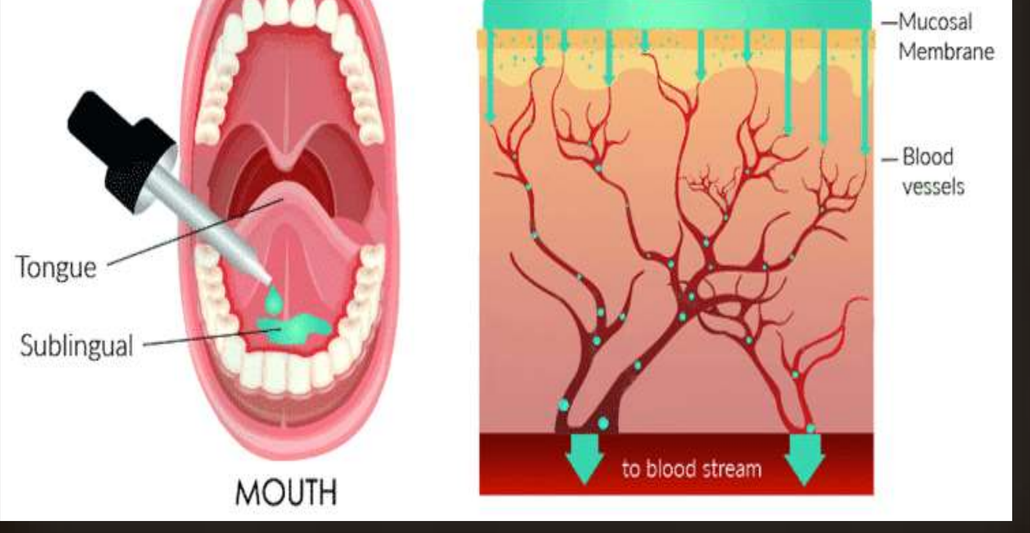
Cannabinoid Drop
Mucosal Membrane
Blood vessels
Tongue
Sublingual
MOUTH
to blood stream

CONCLUSION

CBD oil solutions in different concentrations have been largely used in many countries. These preparations that can be found on our pharmaceutical market are an essential borderline product especially for cancer patients. The importance of technological processes must be emphasized in order to obtain high quality and safety products for our patients.

REFERENCES:

-Jacobs, et al. 2016. Exp. Clin. Psychopharmacol. 24, 320–330.

-Larsen & Shahinas. 2020. J. Clin. Med. Res. 12(3), 129–141.

-Pertwee & Cascio. 2014. Handbook of Cannabis. Oxford Scholarship; Oxford, UK.

-Official Gazette of R.M no. 203/2015. Agency for Medicines and Medical Devices MALMED. Skopje, 2015.

-Official Gazette of R.M no. 37/2016. Ministry of Health. Skopje, 2016.

14th CESPT OHRID, N. MACEDONIA

* Contact: mihail.311155@student.ugd.edu.mk