Reverse phase LC method for in vitro dissolution test for determination of Bromazepam from tablet formulations

Irena Brchina[1], Biljana Gjorgjeska[2]*

[1]AD Jaka 80,

[2]University Goce Delcev, Faculty of Medical Sciences

Introduction

The dissolution tests became an essential parameter to determine the properties of pharmaceutical formulations in order to predict their quality. The quality is directly associated with the patient's health. Thus, there is a real need to develop dissolution tests able to predict in vivo physiological behavior.

Goals

The aim of this study was to develop an analytical method for in vitro dissolution test for determination of Bromazepam in tablet formulations by using HPLC technique and to validate it.

Materials and methods

Tablet dissolution test:
For dissolution medium 0.1 M HCl was chosen, in volume of 500 ml, at 37ºC, performed on ERWEKA DT 700, apparatus 2 (paddle), with 75 rpm for 45 minutes.

Analytical method for content of Bromazepam: High Performance Liquid Chromatography, validation was carried out on Shimadzu Nexera HPLC system.

A satisfactory separation, good peak symmetry and optimal retention time was obtained with mobile phase consisting a mixture of methanol, acetonitrile and potassium dihydrogen phosphate buffer (KH_2PO_4) (pH 7.0; 11.33g/l of KH_2PO_4) in ratio of 45:5:50 (v/v/v) that was set at flow rate of 1.0 ml/min was found to be optimum and further optimized by adjusting pH 7.0 by adding KOH 0.5M. A LiChrospher RP Select B column (125 × 4.0 mm, 5µm) is used as stationary phase with temperature of column oven, 50ºC.

Conclusions

The proposed method is simple, rapid, accurate, precise, and specific without interference of excipients. Its chromatographic run time of 3.50 min allows the analysis of a large number of samples in short period of time. Therefore, it is suitable for the routine analysis of Bromazepam in pharmaceutical dosage forms. So it could be used for the rapid and reliable determination of Bromazepam in tablet formulations.

Results and Discussion

A simple reverse phase HPLC method for in vitro dissolution test was developed and validated for the determination of Bromazepam and release from pharmaceutical dosage form. The elution was monitored at 239 nm. Chromatogram showed a peak of Bromazepam (BZP) at retention time of 3.50 ± 0.1 min. The linearity of Bromazepam was confirmed with correlation coefficient of 0.999. Tipical chromatogram driven from Bromazepam tablets is showen on *Figer 1*. Precision of the peak area response and RT for the peak of Bromazepam is showen on *Figer 2*.

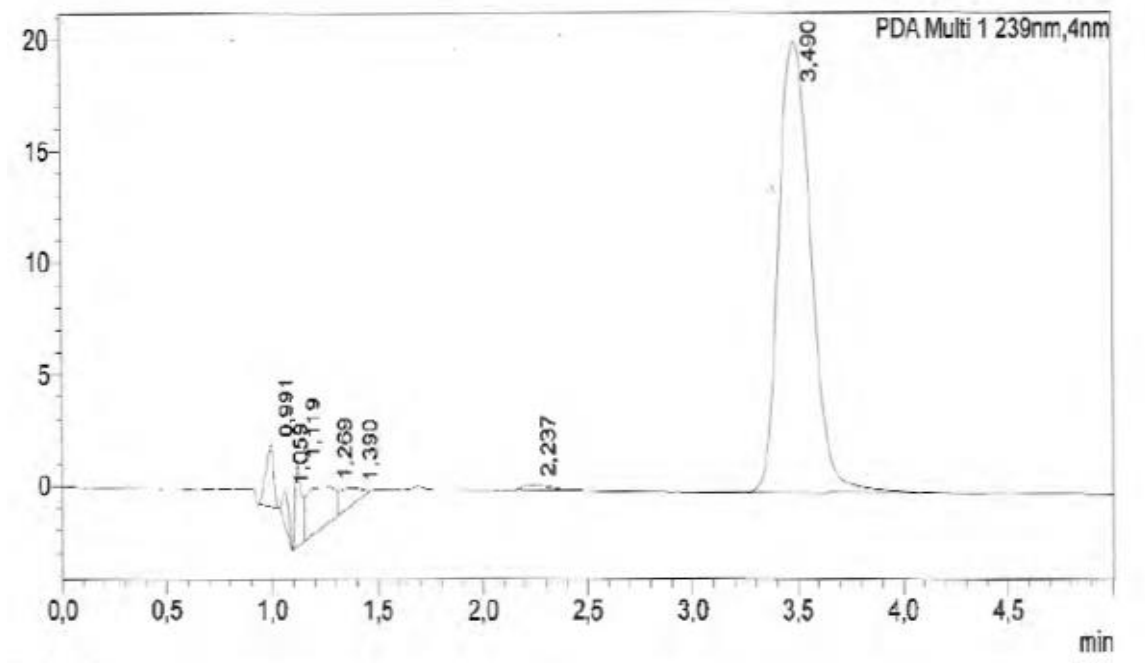

PDA Multi 1 239nm,4nm
3,490
2,237
0,991
1,269
1,390
20
15
10
5
0
0,0
0,5
1,0
1,5
2,0
2,5
3,0
3,5
4,0
4,5
min

Replicates	Precision of the peak area response and RT for the peak of Bromazepam	
	Peak area	Retention time
1	360137	3,505
2	360996	3,506
3	359452	3,523
4	360364	3,522
5	361410	3,505
6	358946	3,492
Mean	360217	3,508
SD	922,90	0,0117
RSD	0,2562	0,3359

The suitability of the mobile phase determined on the basis of the sensitivity of the dissolution, time required for the analysis, easy way of preparation and use of readily available cost effective solvents are benefits of the proposed method. The method was validated as per ICH guidelines with respect to specificity, linearity, accuracy, precision, robustness, solution stability and filter paper compatibility. All results of validation parameters meet the limits of ICH guidelines-(ICH Q2, 2005)